Developing and Implementing Key Performance Indicators (KPI) for Endovascular Thrombectomy in Ontario

Authors: Kathryn Yearwood, Dr. Richard H. Swartz, Anar Pardhan, Phongsack Manivong, Julie Tang, Leah Justason, Shelley A. Sharp, Dr. Mark Bayley, Dr. Leanne K. Casaubon, Kathy Godfrey, Dr. Moira K. Kapral, Elizabeth Linkewich, Rhonda McNicoll-Whiteman, Joan Porter, Dr. Grant Stotts, Dr. Amy Y. X. Yu, Mirna Rahal

CorHealth Ontario
Advancing cardiac, stroke and vascular care

Background

In Ontario, Endovascular Thrombectomy (EVT) is performed at 10 specialized hospitals across the province (Figure 1). In 2017, the Ministry of Health and Long-Term Care (MOHLTC) requested that CorHealth Ontario[1] establish a framework for measuring, monitoring and reporting on EVT performance to ensure alignment with best practices, improve system planning and drive quality/system improvements. To support the development of this framework CorHealth Ontario leveraged the expertise of the provincial EVT Performance Measurement and Monitoring (EVT PMM) Task Group. This group reports to CorHealth Ontario's EVT Steering Committee as part of CorHealth Ontario's external governance structure and consists of a group of content experts, including neurologists, clinical nurse specialists, epidemiologists and other key stakeholders with expertise in stroke system evaluation and implementation.

Figure 1. Map of hospitals in Ontario that offer hyperacute stroke care.

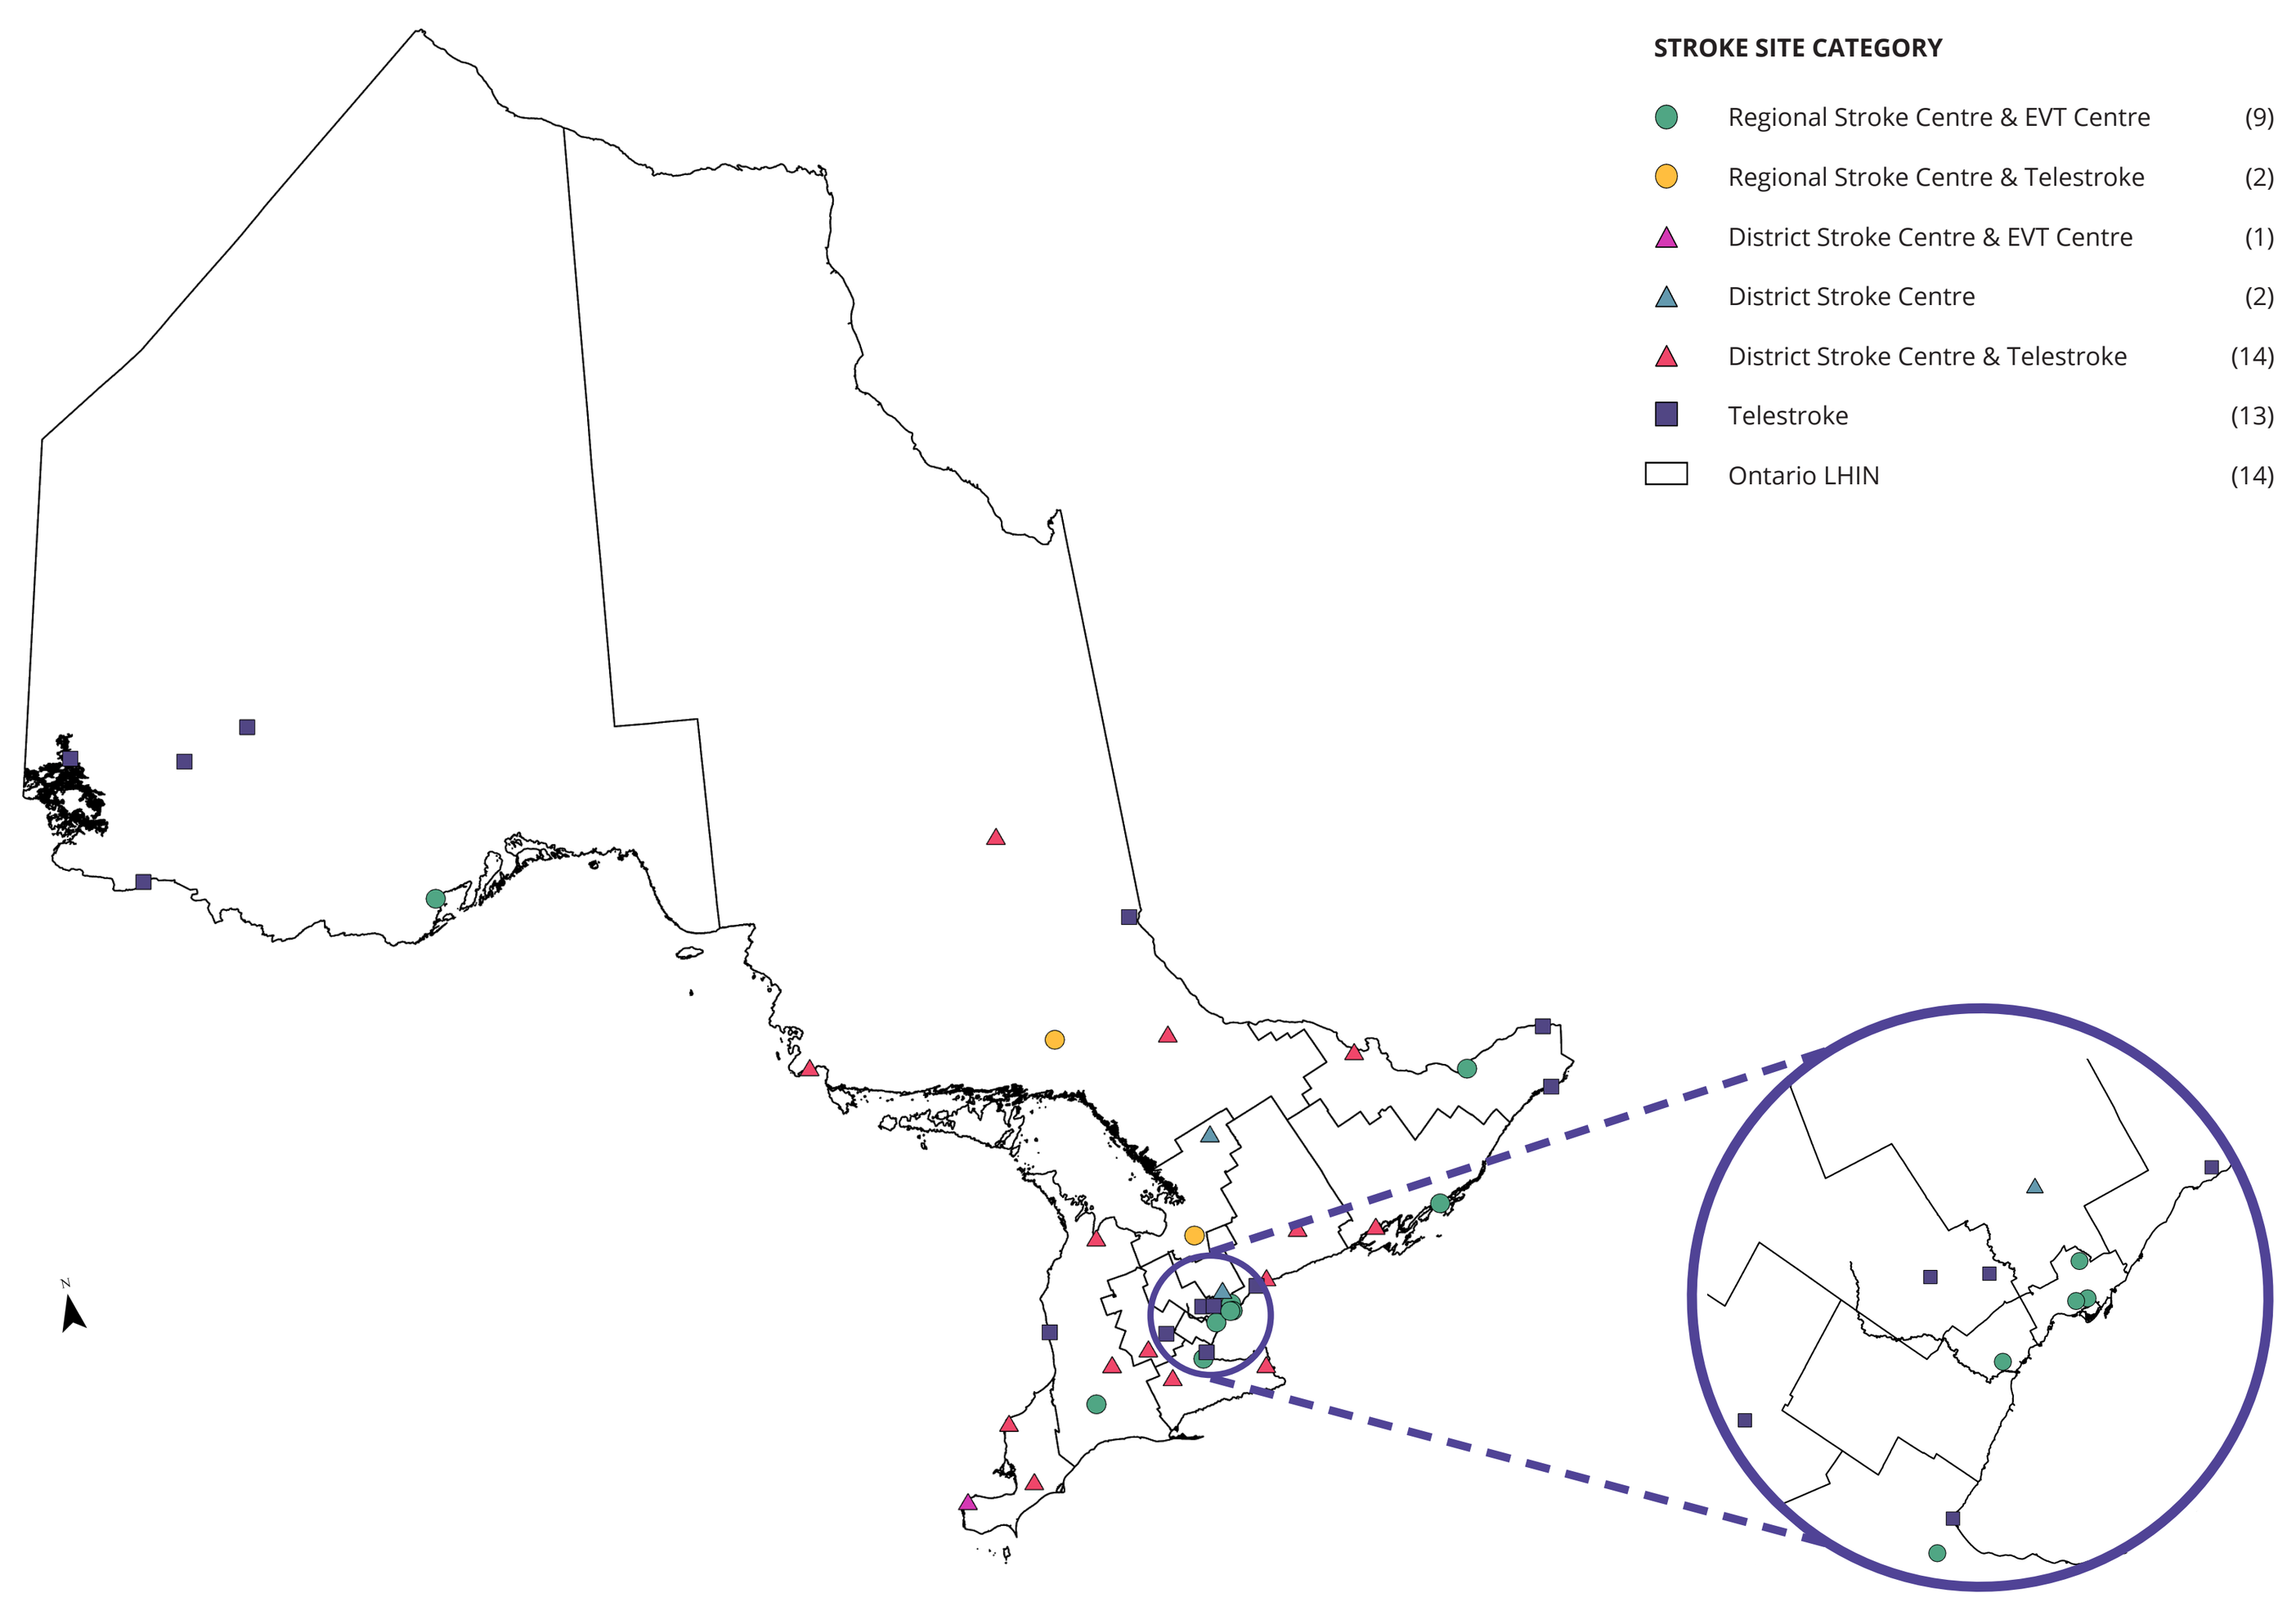
STROKE SITE CATEGORY
Regional Stroke Centre & EVT Centre (9)
Regional Stroke Centre & Telestroke (2)
District Stroke Centre & EVT Centre (1)
District Stroke Centre (2)
District Stroke Centre & Telestroke (14)
Telestroke (13)
Ontario LHIN (14)

Methods

EVT INDICATOR SELECTION

The task group identified 22 indicators and prioritized these for reporting based on the following considerations:

- **Level of Reporting:** patient outcomes, access, system performance and key processes
- **Data Availability:** ability to calculate the indicator using existing data sources
- **Redundancy:** whether the indicator is captured in other provincial reports
- **Distribution of Indicators:** where the indicator fits in the patient journey – pre EVT, during EVT and post EVT
- **Quality Domain:** captures various aspects of quality including timeliness, equity, and effectiveness

Of the 22 proposed indicators, 12 could be calculated using existing data held by the Canadian Institute for Health Information (CIHI). After factoring in the level of reporting, redundancy, distribution and quality of these indicators, 3 were excluded, resulting in a total of 9 key performance indicators.

DEFINING THE COHORT AND DEVELOPMENT OF TECHNICAL SPECIFICIATIONS

- Iteratively developed by CorHealth Ontario's analytics team in collaboration with the EVT PMM Group
- ICES, as an independent and experienced entity in health services research, reviewed indicator technical specifications and provided recommendations for further refinement
- CorHealth Ontario's analytics team calculated all indicators and vetted results through the Regional Stroke Program Managers/Directors at the EVT hospitals to ensure alignment between results and individual tracking/clinical experience

DEVELOPMENT OF A REPORTING TOOL

- Internal blue-sky thinking session to develop a vision for the report
 - Key takeaways: report should "tell a story"
- "The story" is told through four questions (Table 1)
 - Each indicator is aligned with a question
- Mock-up of dashboard created, and feedback obtained from key stakeholder (e.g. Regional Stroke Program Managers/Directors)
- Integrated Decision Support (IDS), a technology infrastructure hosted and delivered by Hamilton Health Sciences, created and implemented an interactive dashboard of indicator results and made it available to all EVT sites (Figure 2)

Results

1. QUALITY DOMAINS/QUESTIONS AND KEY PERFORMANCE INDICATORS

Table 1. Quality domains/questions and key performance indicators

QUALITY DOMAINS/QUESTIONS	INDICATORS
TIMELINESS: Are patients being identified and treated in a timely manner?	• Median time from emergency department (ED) arrival (at EVT site) to qualifying computed tomography angiograph (CTA), computed tomography perfusion (CTP), magnetic resonance angiography (MRA) • Median time from ED arrival (at EVT site) to arterial puncture • Median time from ED arrival (at EVT site) to time of first reperfusion
EQUITY: Do patients have equitable access to EVT throughout the province?	• Proportion and number of ischemic stroke patients who receive an EVT procedure (cross-regional comparison in detailed pages)
EFFECTIVENESS: Are the appropriate patients being identified, referred, and accepted for EVT?	• Proportion of patients transferred to an EVT centre for EVT who received EVT procedure by LHIN and/or facility
EFFECTIVENESS: Are the desired outcomes being achieved?	• 30-day risk-adjusted all-cause mortality rates for patients who received EVT • Median number of days EVT patients spend at home in the first 90 days post procedure • Proportion of EVT patients successfully reperfused

2. REPORTING TOOL: EVT DASHBOARD

The EVT Dashboard landing page (Figure 2) provides users with a visual representation of the EVT patient journey:

- Users have the option to view either provincial or hospital-specific results
- Each indicator is linked to a more detailed page which focuses on one of the quality domains/questions and provides related indicator results

The detailed pages (Figure 2) present results graphically and allow for cross-hospital comparison:

- Where available, targets are included
- Denominator and numerator values, and 25th, 75th and 50th percentiles are available for each hospital in the province
- Outcome indicators are stratified to include/exclude in-hospital strokes

Figure 2. EVT Dashboard landing page indicating the ability to click on specific sections for more detailed pages, including hospital level results.

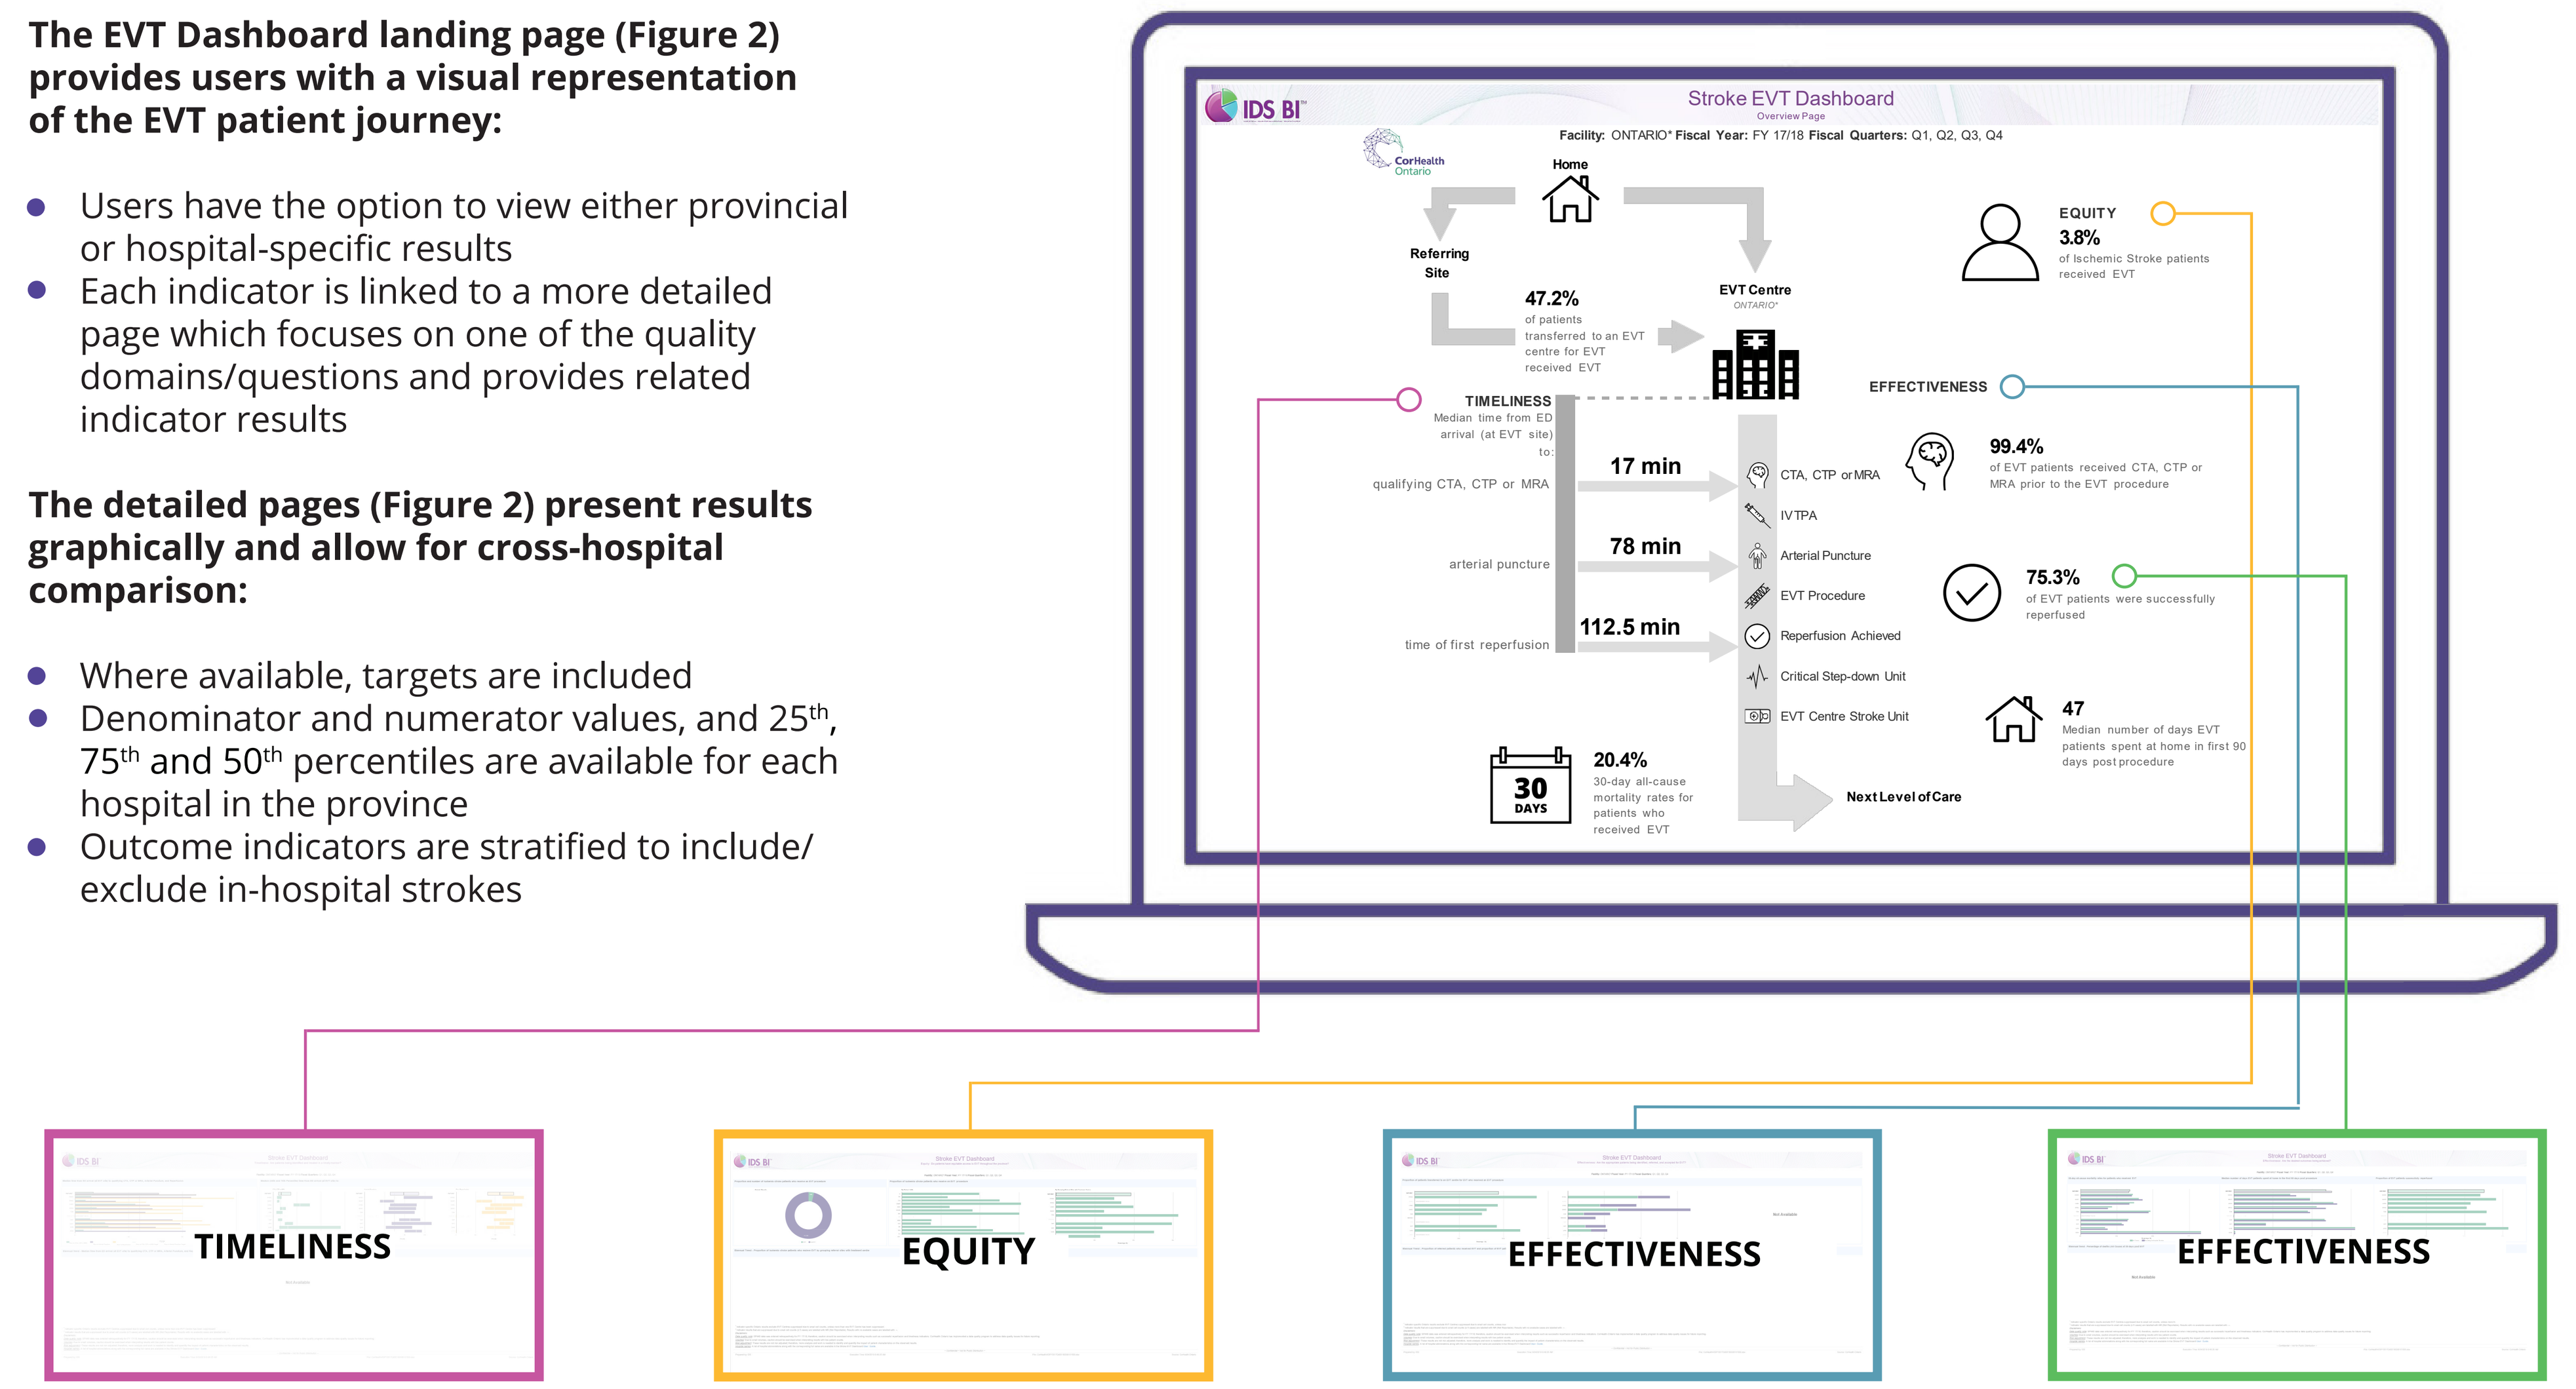
IDS BI
Stroke EVT Dashboard
Overview Page
Facility: ONTARIO* Fiscal Year: FY 17/18 Fiscal Quarters: Q1, Q2, Q3, Q4
Home
Referring Site
EVT Centre
47.2%
of patients transferred to an EVT centre for EVT received EVT
EQUITY
3.8%
of Ischemic Stroke patients received EVT
EFFECTIVENESS
TIMELINESS
Median time from ED arrival (at EVT site) to:
qualifying CTA, CTP or MRA
17 min
arterial puncture
78 min
time of first reperfusion
112.5 min
CTA, CTP or MRA
IVTPA
Arterial Puncture
EVT Procedure
Reperfusion Achieved
Critical Step-down Unit
EVT Centre Stroke Unit
99.4%
of EVT patients received CTA, CTP or MRA prior to the EVT procedure
75.3%
of EVT patients were successfully reperfused
47
Median number of days EVT patients spent at home in first 90 days post procedure
30 DAYS
20.4%
30-day all-cause mortality rates for patients who received EVT
Next Level of Care
TIMELINESS
EQUITY
EFFECTIVENESS
EFFECTIVENESS

3. QUALITY IMPROVEMENT

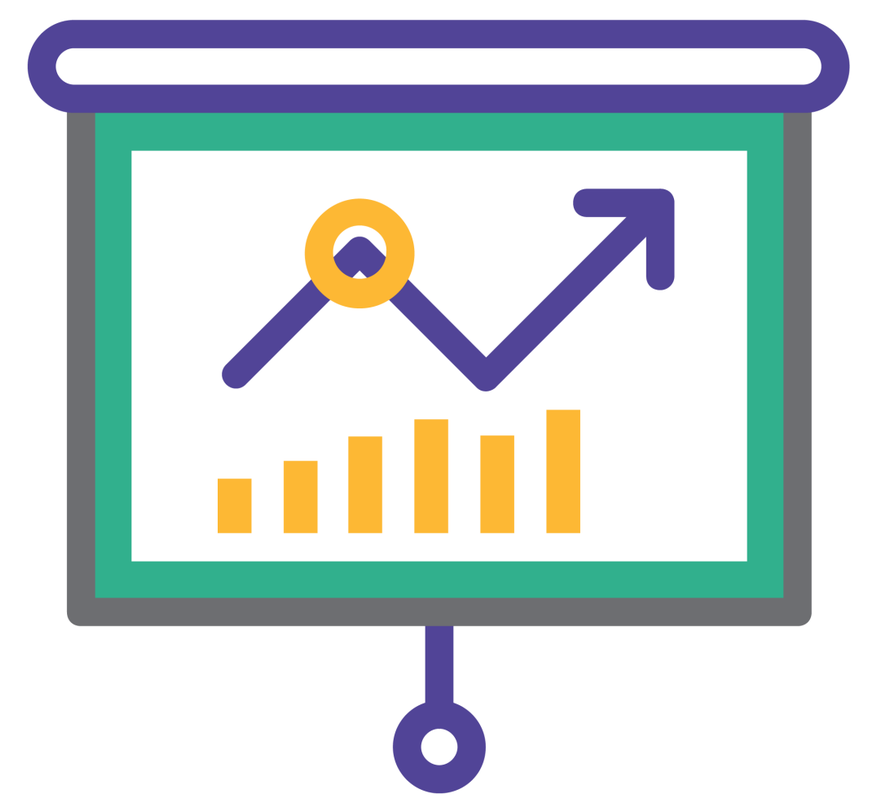

PROVINCIAL QUALITY IMPROVEMENT

Access to EVT varies considerably between patients presenting directly to an EVT hospital and those who first present to a hospital that only provides thrombolysis (i.e. tissue plasminogen activator (tPA)) for stroke. As such, the EVT Steering Committee has requested the EMS/Patient Transport Task Group to develop recommendations regarding the use of large vessel occlusion (LVO) screening tools with Emergency Medical Service Providers to streamline access to EVT hospitals for those patients likely to be eligible for EVT.

LOCAL QUALITY IMPROVEMENT

In September 2019, biannual and annual trending will be included for each indicator to enable individual hospitals to track their progress over time.

4. DATA QUALITY (DQ)

CorHealth Ontario developed a data quality (DQ) management model, the DQ and Compliance Program, to ensure data is of high-quality and fit-for-use, with a focus on improving data at the source.

The DQ and Compliance Program was applied to EVT data and consists of the following:

- 9 EVT DQ indicators which were identified and validated by the EVT PMM Task Group
- A report showing the indicators compared against thresholds/acceptable values
- A quarterly process for review and feedback

Hospitals use the report to investigate, correct issues, provide feedback to CorHealth Ontario, and put in place data improvement plans to resolve issues and help prevent them from reoccurring.

The DQ and Compliance Program has increased the quality of the EVT data. With this increased quality comes increased confidence in reporting and the ability to create accurate action plans, all with the goal of improving EVT services in Ontario.

Conclusion

The key performance indicators (KPIs) and reporting process mark a critical milestone in promoting successful implementation of EVT in Ontario. These indicators are intended to drive quality practice improvement and inform system planning at institution and population levels.

Next Steps

- CorHealth Ontario to develop a provincial quality improvement process for EVT that will leverage these results
- Performance of individual hospitals, based on key performance indicators, may be used to inform future funding recommendations
- Integrate the EVT KPI into other stroke reporting to illustrate the complete picture of stroke care in Ontario
- Explore the opportunity to obtain additional information from the referral hospitals to better understand patient outcomes

Acknowledgements

CorHealth Ontario would like to extend their gratitude to the following for their involvement in this project:

- Integrated Decision Support (IDS)
- Lindsay Arscott and Lindsay Siurna, Data Quality, CorHealth Ontario
- Richelle Himaya, Design, CorHealth Ontario
- ICES

References

1. CorHealth Ontario is a central agency that advises the Ministry of Health and Long-Term Care (public funder), hospitals, and care providers to improve the quality, efficiency, accessibility and equity of cardiac, stroke and vascular services for patients across Ontario.